Physicochemical Stability of Chloral Hydrate 25 mg/mL and 100 mg/mL Oral Suspensions in SuspendIt®

Kendice Ip*, Maria Carvalho, Fabiana Vieira-Banov and Daniel Banov
PCCA, Professional Compounding Centers of America, Houston (TX)

Introduction

Chloral hydrate is a hypnotic, sedative and anxiolytic (non-benzodiazepine) drug that is commonly administered to children prior to painless procedures (e.g. dental sedation) or in cases of insomnia for short-term use only (1). Due to a lack of age-appropriate commercial medicines, an alternative extemporaneous preparation was developed for chloral hydrate 25 mg/mL and 100 mg/mL in the oral vehicle SuspendIt® (sugar-free, paraben-free, and gluten-free). The corresponding Beyond-Use Dates (BUDs) were determined by testing the physicochemical stability of the oral suspensions, and were complemented by an in-use stability testing.

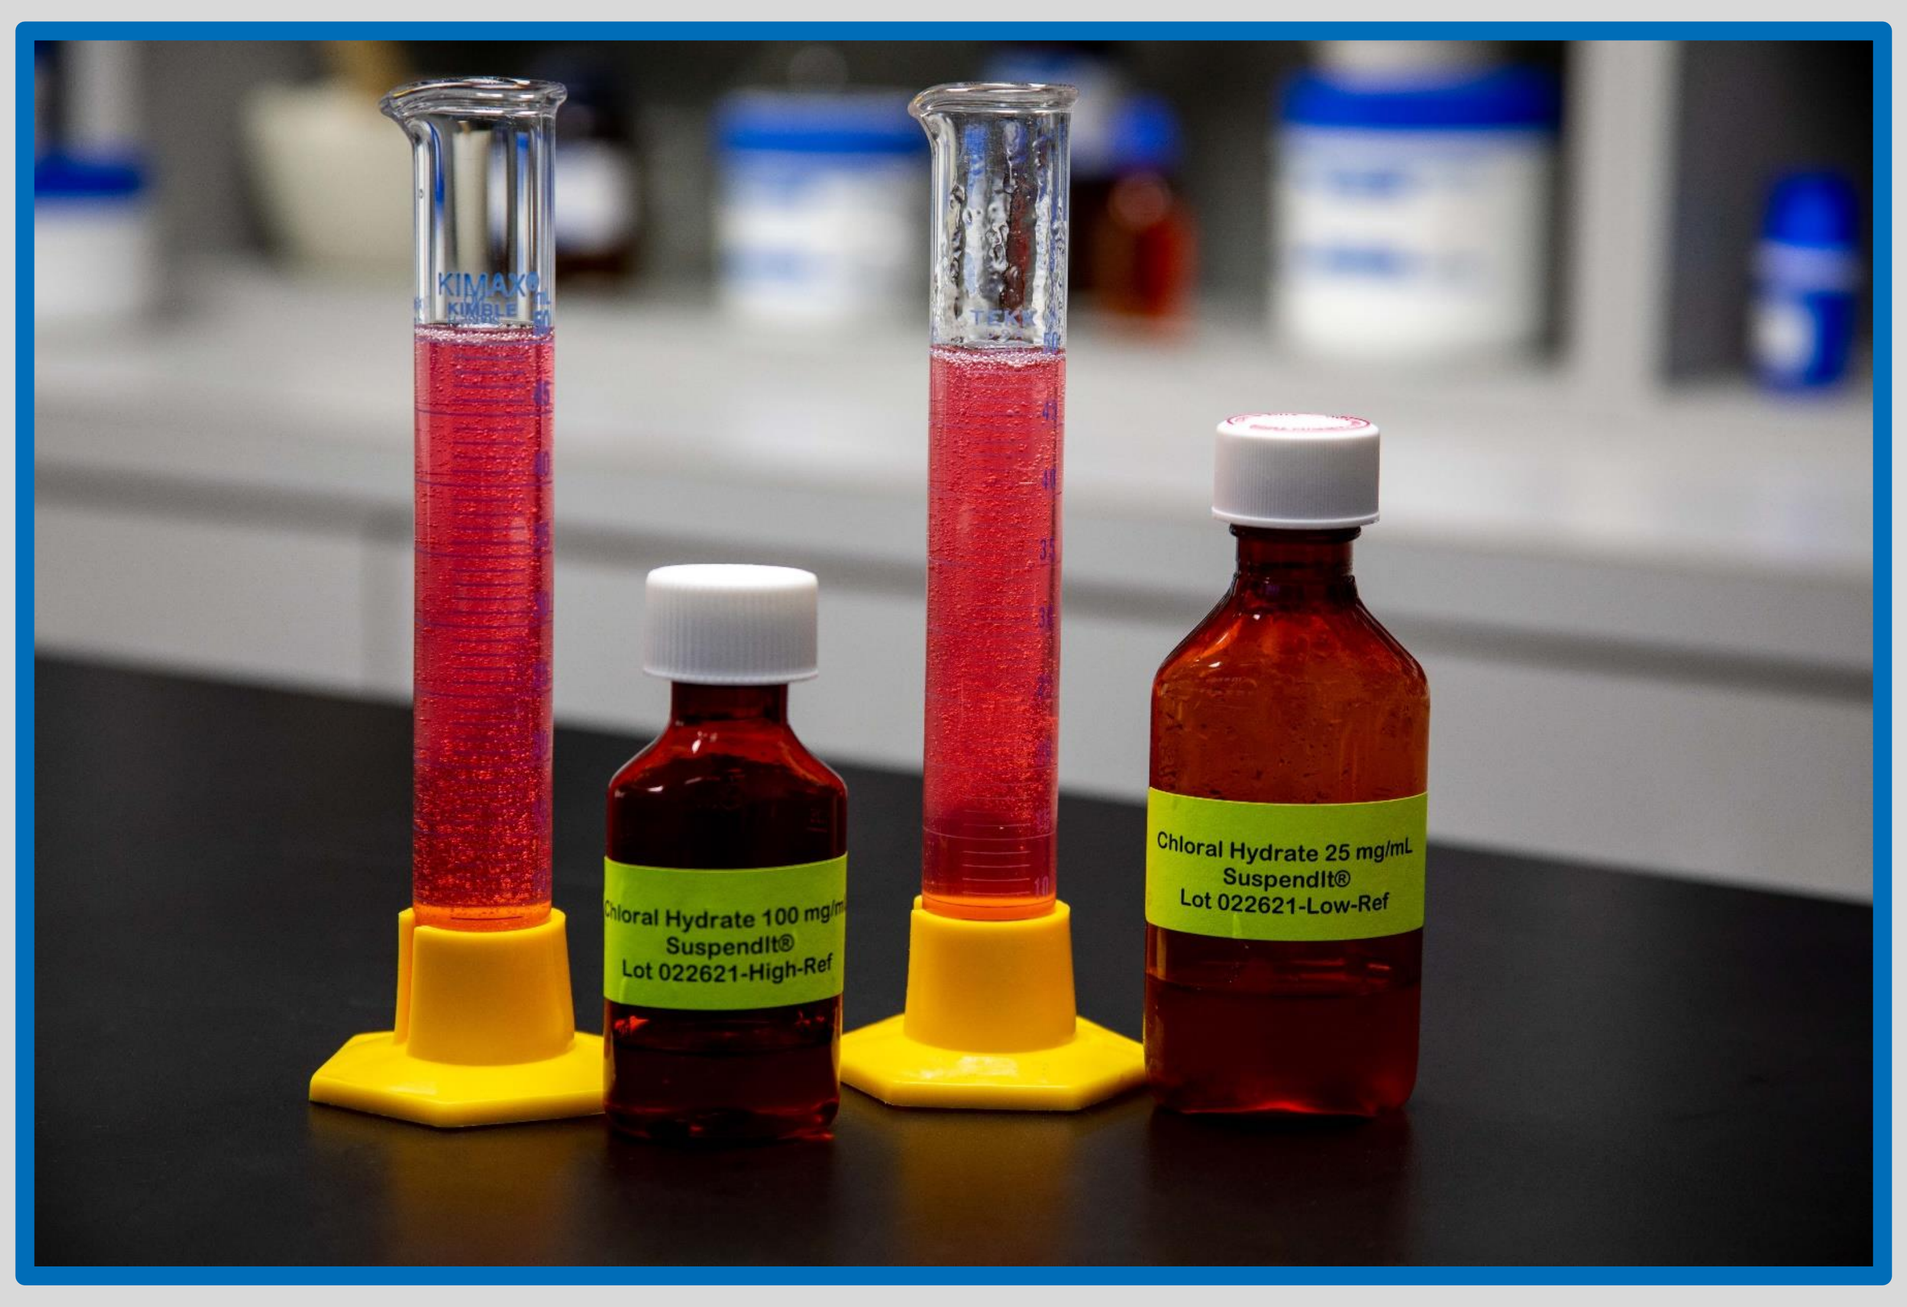

Figure 1. *Samples of chloral hydrate 25 mg/mL (right) and 100 mg/mL (left) on day 180 of the study, at refrigerated temperature.*

Methodology

Two batches of 480 mL were prepared for chloral hydrate 25 mg/mL and also for chloral hydrate 100 mg/mL by adding the active ingredient to SuspendIt®, as well as the following excipients: acesulfame potassium, steviol glycosides 95%, sweetener (Magnasweet 135®), and flavors (raspberry and marshmallow).

Each oral suspension was distributed into amber plastic bottles and the study samples were then stored for 180 days in a laboratory refrigerator at a temperature of 5^0C ± 3^0C; and in an environmentally controlled chamber at a temperature of 25^0C ± 2^0C and relative humidity of 60% ± 5%. One of the batches for chloral hydrate 100 mg/mL was set aside to evaluate the in-use stability of the oral suspensions. As such, these bottles were removed from the storage conditions and uncapped once a day, for the total duration of the study, in order to mimic the patients' daily administration of the oral suspension. At pre-determined time points [0 (baseline), 14, 31, 60, 90 and 180 days], a study sample (one unopened bottle and the daily uncapped bottle) of each strength was withdrawn from the storage conditions, shaken vigorously and tested for physicochemical stability (Figure 1). The physical characterization consisted in observing all samples for appearance/color and odor, and testing for pH (SevenCompact S220 pH Meter; Mettler Toledo) and density (Density Excellence D4 density meter; Mettler Toledo). The chemical characterization consisted in a modified USP titration assay method (2), in which the end-point was indicated by a visible color change. The physicochemical measurements were done in triplicate for all tests.

Results & Discussion

Considering the physical characterization, the chloral hydrate oral suspensions stored at both temperatures, including the in-use testing, exhibited a homogeneous bright pink color (Figure 2, left), a translucent smooth appearance and a distinct sweet marshmallow odor throughout the complete study. For the chloral hydrate 25 mg/mL oral suspensions, the range of densities (1.0145 - 1.0176 g/mL at 5°C and 1.0156 - 1.0174 g/mL at 25°C) and the range of pH (4.85-4.88 at 5°C and 4.77-4.89 at 25°C) were within the limits. For the chloral hydrate 100 mg/mL oral suspensions, the range of densities and pH were also within the limits, for both stability and in-use testing: 1.0504-1.0522 g/mL and 4.88-4.93 at 5°C; 1.0506-1.0531 g/mL and 4.66-4.90 at 25°C. Considering the chemical characterization, each titration was indicated by a color change from bright pink to pale pink (Figure 2, left to right). The percent potency was calculated taking into account the baseline measurements on day 0. The potency of the oral suspensions remained within the specifications of ±10% for the duration of the study (3), as follows: 91.4% - 97.0% at 5°C and 95.6% - 97.9% at 25°C (chloral hydrate 25 mg/mL); and 95.1% - 102.7% at 5°C and 94.7% - 101.7% at 25°C (chloral hydrate 100 mg/mL). As a result, the chloral hydrate oral suspensions from 25 mg/mL up to 100 mg/mL (bracketed study) are physically and chemically stable at 5°C and 25°C, in amber plastic bottles, for 180 days.

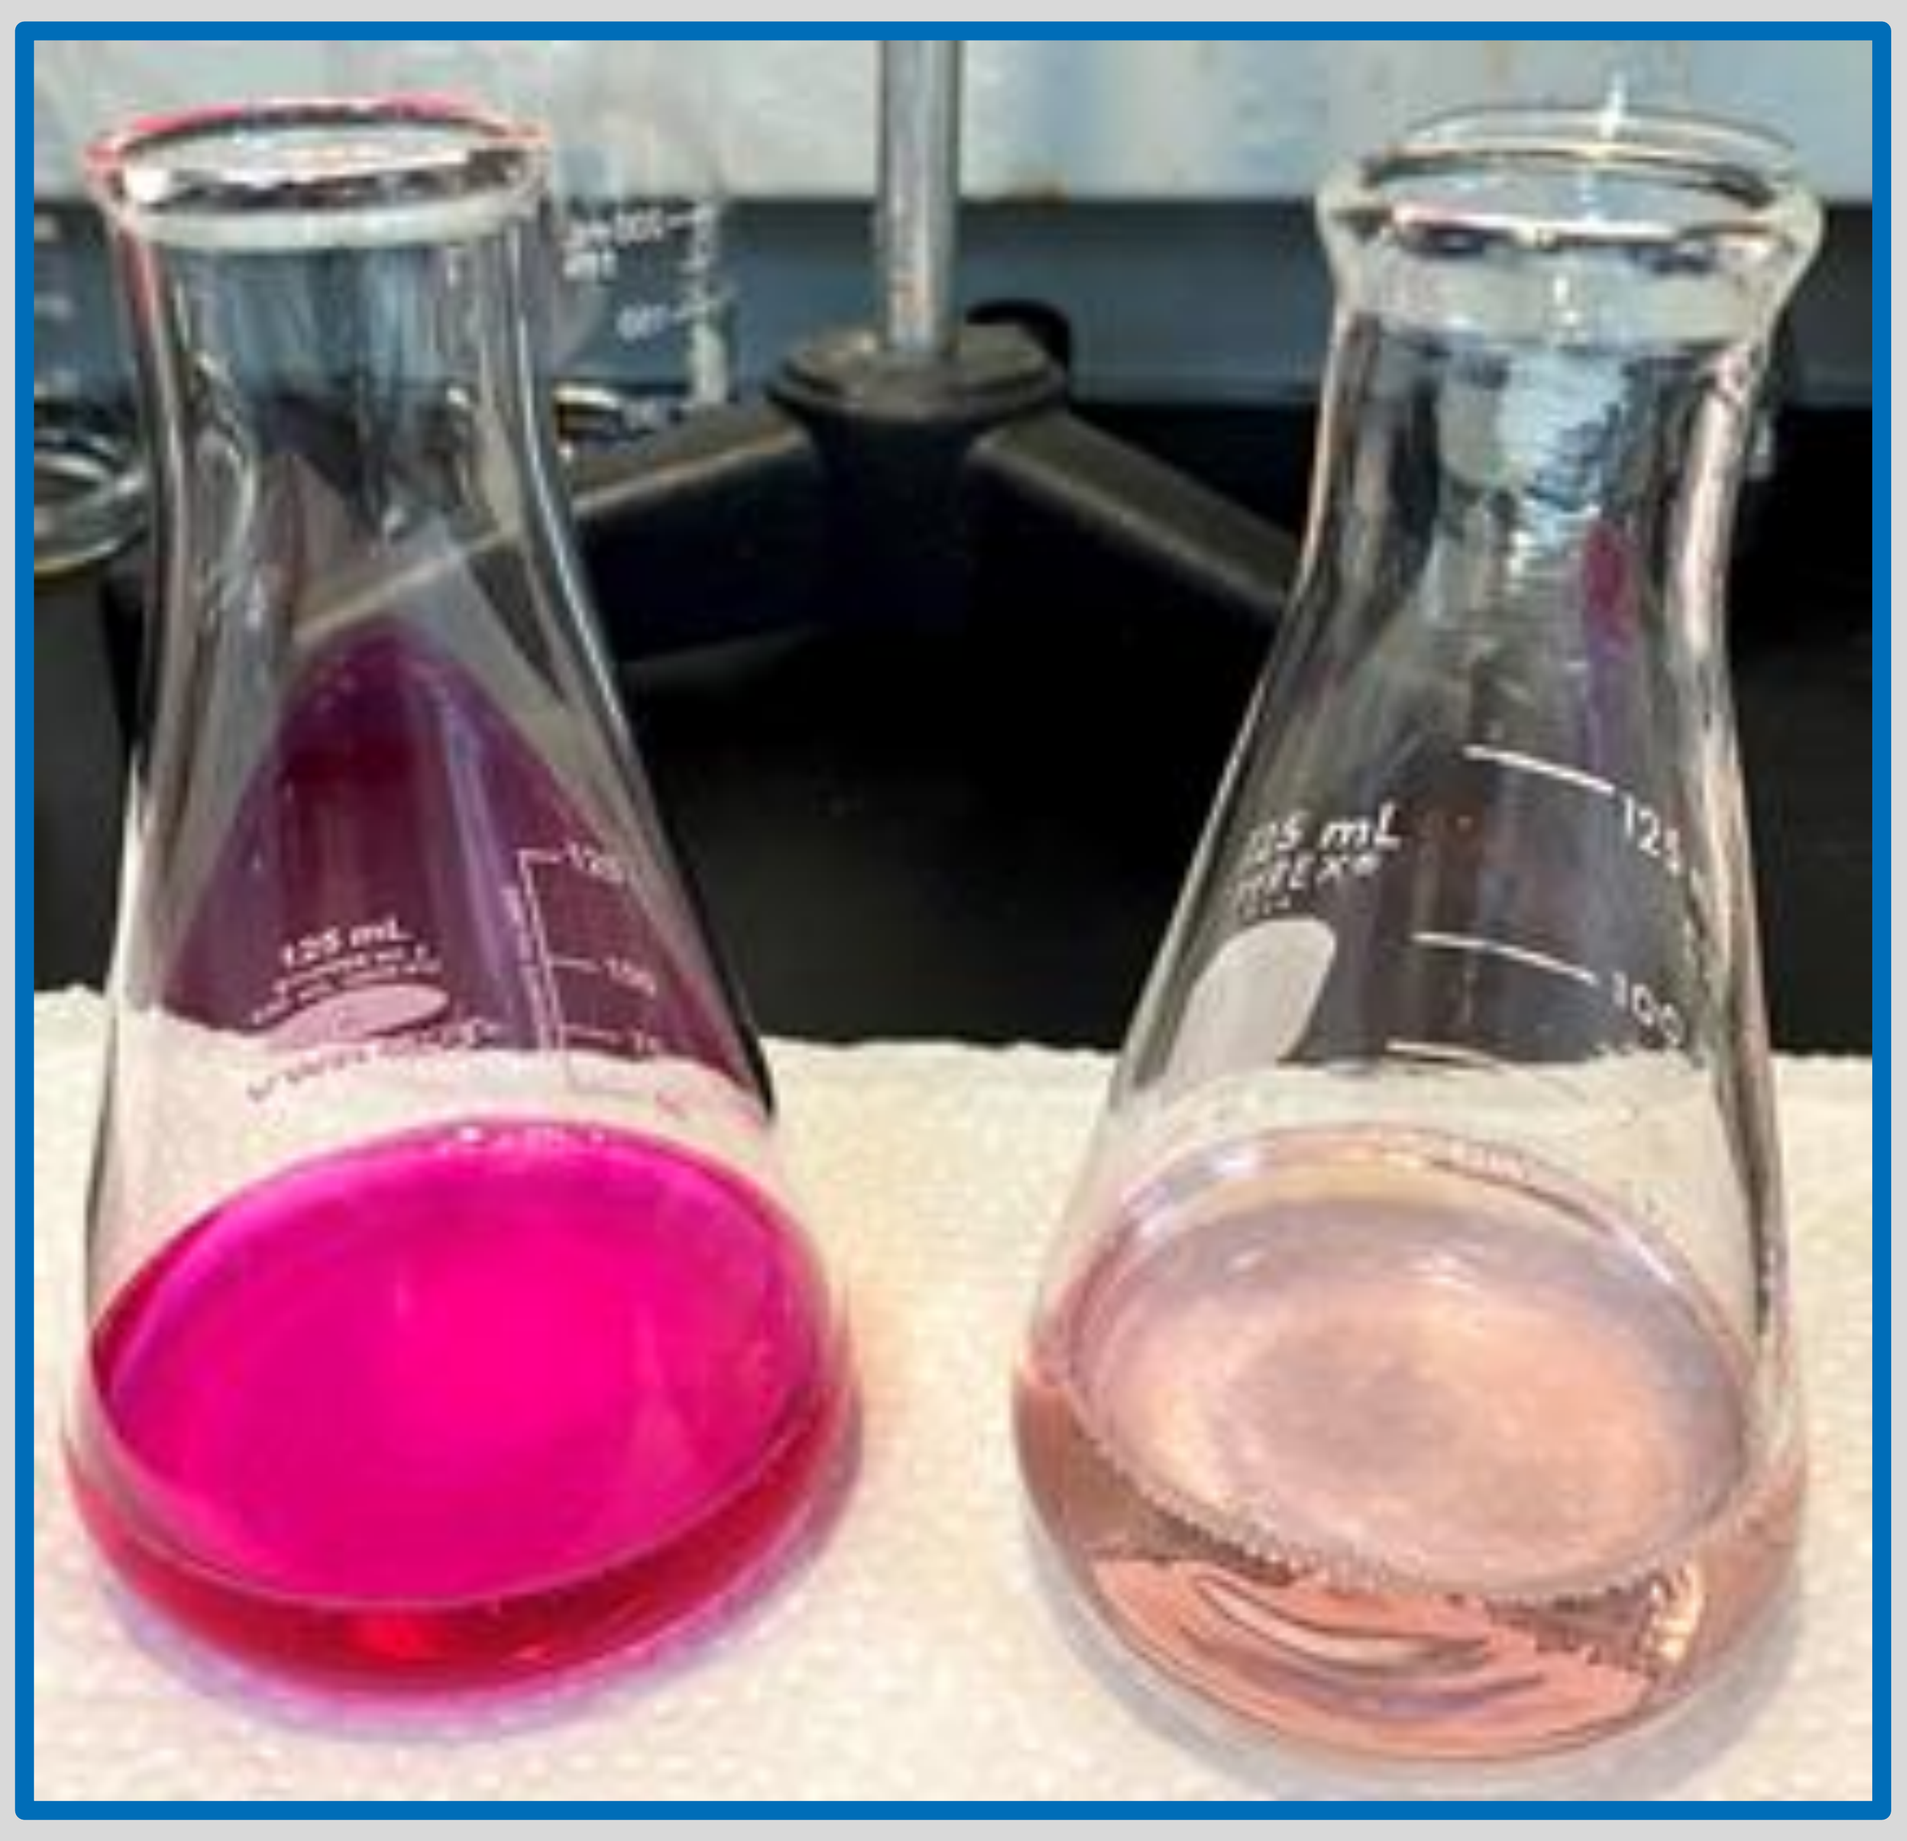

Figure 2. *Before (left) and after (right) titration of the chloral hydrate 25 mg/mL oral suspension on day 90.*

The BUD of the chloral hydrate 25 mg/mL - 100 mg/mL oral suspensions (SuspendIt®) is 6 months at both refrigerated and room temperature, in amber plastic bottles. This BUD was reinforced by the complementary method of in-use stability testing.

References

(1) Paediatric Formulary Committee (2020) *BNF for Children.* London: BMJ Group and Pharmaceutical Press, p.311-2. (2) The United States Pharmacopeial Convention (2021) 'Monographs – Chloral Hydrate Oral Solution'. *USP – NF Online*. Accessed: July 7^{th}, 2021. (3) The United States Pharmacopeial Convention (2016) 'General Requirements / <2> Oral Drug Products – Product Quality Tests'. *USP 39 – NF 34*. Rockville: USP, p.77.

Poster digital version available at: https://www.pccarx.com/Science

**Corresponding author: kip@pccarx.com*